[P0879] The Totality of Evidence for FYB202 – an EU-approved and US-licensed Biosimilar to Reference Ustekinumab

Sigrid Balser, Brigitte Freudensprung, Katharina Huebner, Inga Moreau, Katrin Nopora, Susanne Pippig, Maximilian Schmid, Michael Trieb, Florian Wolschin

Formycon AG, Martinsried/Planegg, Germany

Introduction

- Biosimilars allow more patients access to affordable treatment options and help reduce the financial burden on healthcare systems.[1]
- Ustekinumab is an interleukin (IL)-12 and IL-23 antagonist approved for the treatment of moderate-to-severe plaque psoriasis, active psoriatic arthritis, and active inflammatory bowel disorders.[2]
- FYB202 has been approved by both EMA and FDA in September 2024 as a biosimilar to reference ustekinumab based on extensive comparative analytical characterization data and the proof of equivalence in clinical studies.
- This poster summarizes the totality of evidence proofing similarity in terms of functional assays, pharmacokinetic profiles, as well as clinical performance in patients with moderate-to-severe plaque psoriasis.

Methods

- A full set of critical quality attributes was investigated with a comprehensive set of analytical methods.
- Functional comparability of FYB202 to the reference product (RP) for the main mechanism of action was assessed with IL-12 and IL-23 binding and bioassays.
- Comparable FcRn binding kinetics could be demonstrated for FYB202 and reference ustekinumab.
- Equivalence in PK was shown in a 3-way study comparing FYB202 to EU-approved and to US-licensed reference ustekinumab in 491 healthy volunteers receiving a single subcutaneous dose of 45mg.
 - Primary endpoints: $AUC_{0\text{-inf}}$ and C_{max}
 - Bioequivalence margins: 80 – 125%
 - Secondary endpoints: further PK parameters, immunogenicity, tolerability, and safety
- Equivalent efficacy as well as similarity in safety, tolerability, and immunogenicity were evaluated in a comparative clinical trial in patients with moderate-to-severe plaque psoriasis (NCT04595409)
 - Primary efficacy endpoint: percent improvement in PASI score from baseline to week 12
 - Equivalence criteria:
 - EU-specific: 95% CI to be within (-11%; 11%)
 - US-specific: 90% CI to be within (-10%; 10%)
 - Secondary endpoints: further efficacy endpoints, safety, and immunogenicity
- Serum ustekinumab concentrations were determined by an ELISA with an analytical range of 40 – 1400 ng/mL. Anti-drug antibodies (ADA) were detected using a bridging ECLIA in a 3-tier approach, followed by neutralizing ADAs (NAbs) determination in a competitive ligand-binding ELISA. For all assays a one-assay approach was established, using one assay for the measurement of reference product and biosimilar concentrations and ADAs for best comparison. The same assays were used in all clinical trials.

Results

- FYB202 was found to be **analytically highly similar** to reference ustekinumab.
- The most relevant potency results, which are relevant for the mechanism of action of ustekinumab, are shown in Fig. 1.

Figure 1: Potency assays addressing IL-12 and IL-23 binding

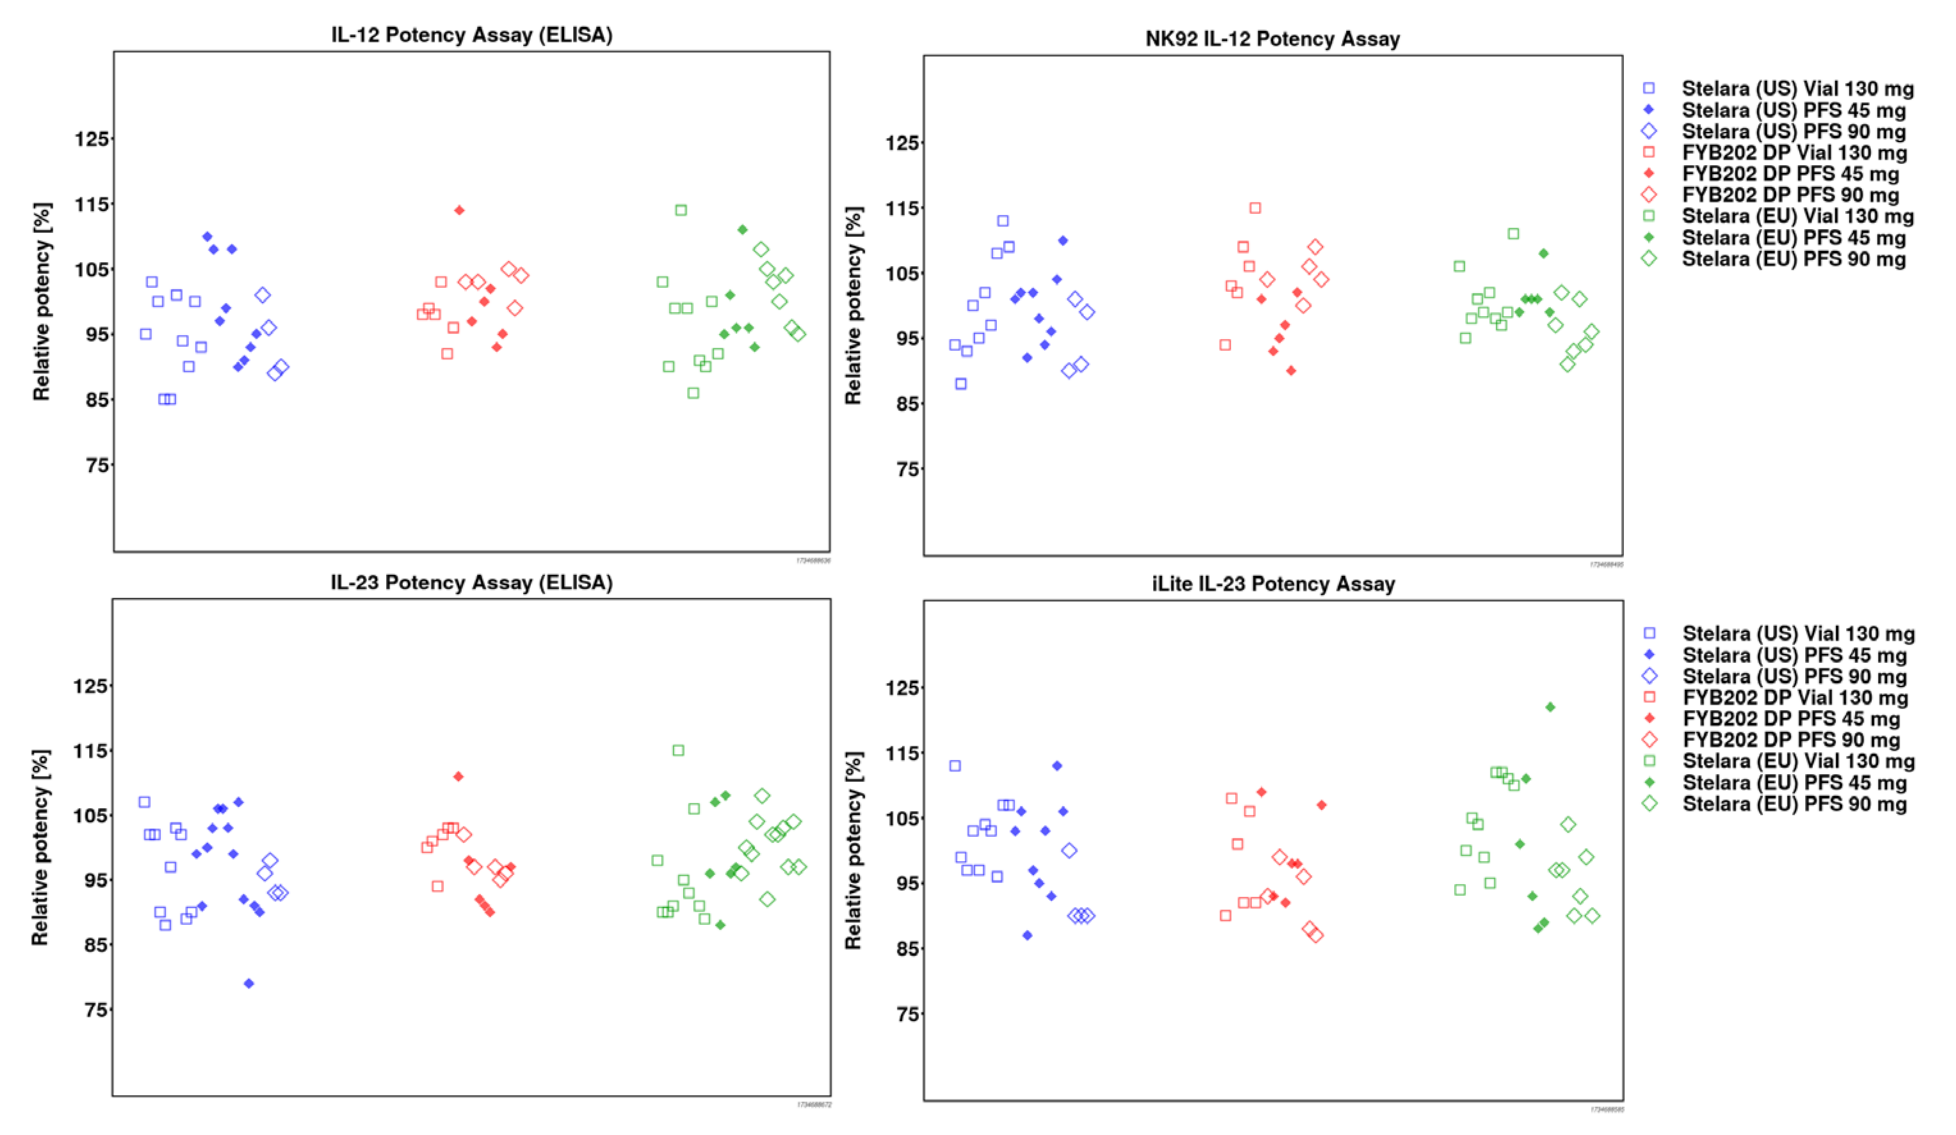
IL-12 Potency Assay (ELISA)
NK92 IL-12 Potency Assay
IL-23 Potency Assay (ELISA)
iLite IL-23 Potency Assay
Relative potency [%]
Stelara (US) Vial 130 mg
Stelara (US) PFS 45 mg
Stelara (US) PFS 90 mg
FYB202 DP Vial 130 mg
FYB202 DP PFS 45 mg
FYB202 DP PFS 90 mg
Stelara (EU) Vial 130 mg
Stelara (EU) PFS 45 mg
Stelara (EU) PFS 90 mg

- Results for FcRn binding, which is important for the half-life of the molecule, are shown in Fig. 2.

Figure 2: FcRn Binding Kinetics

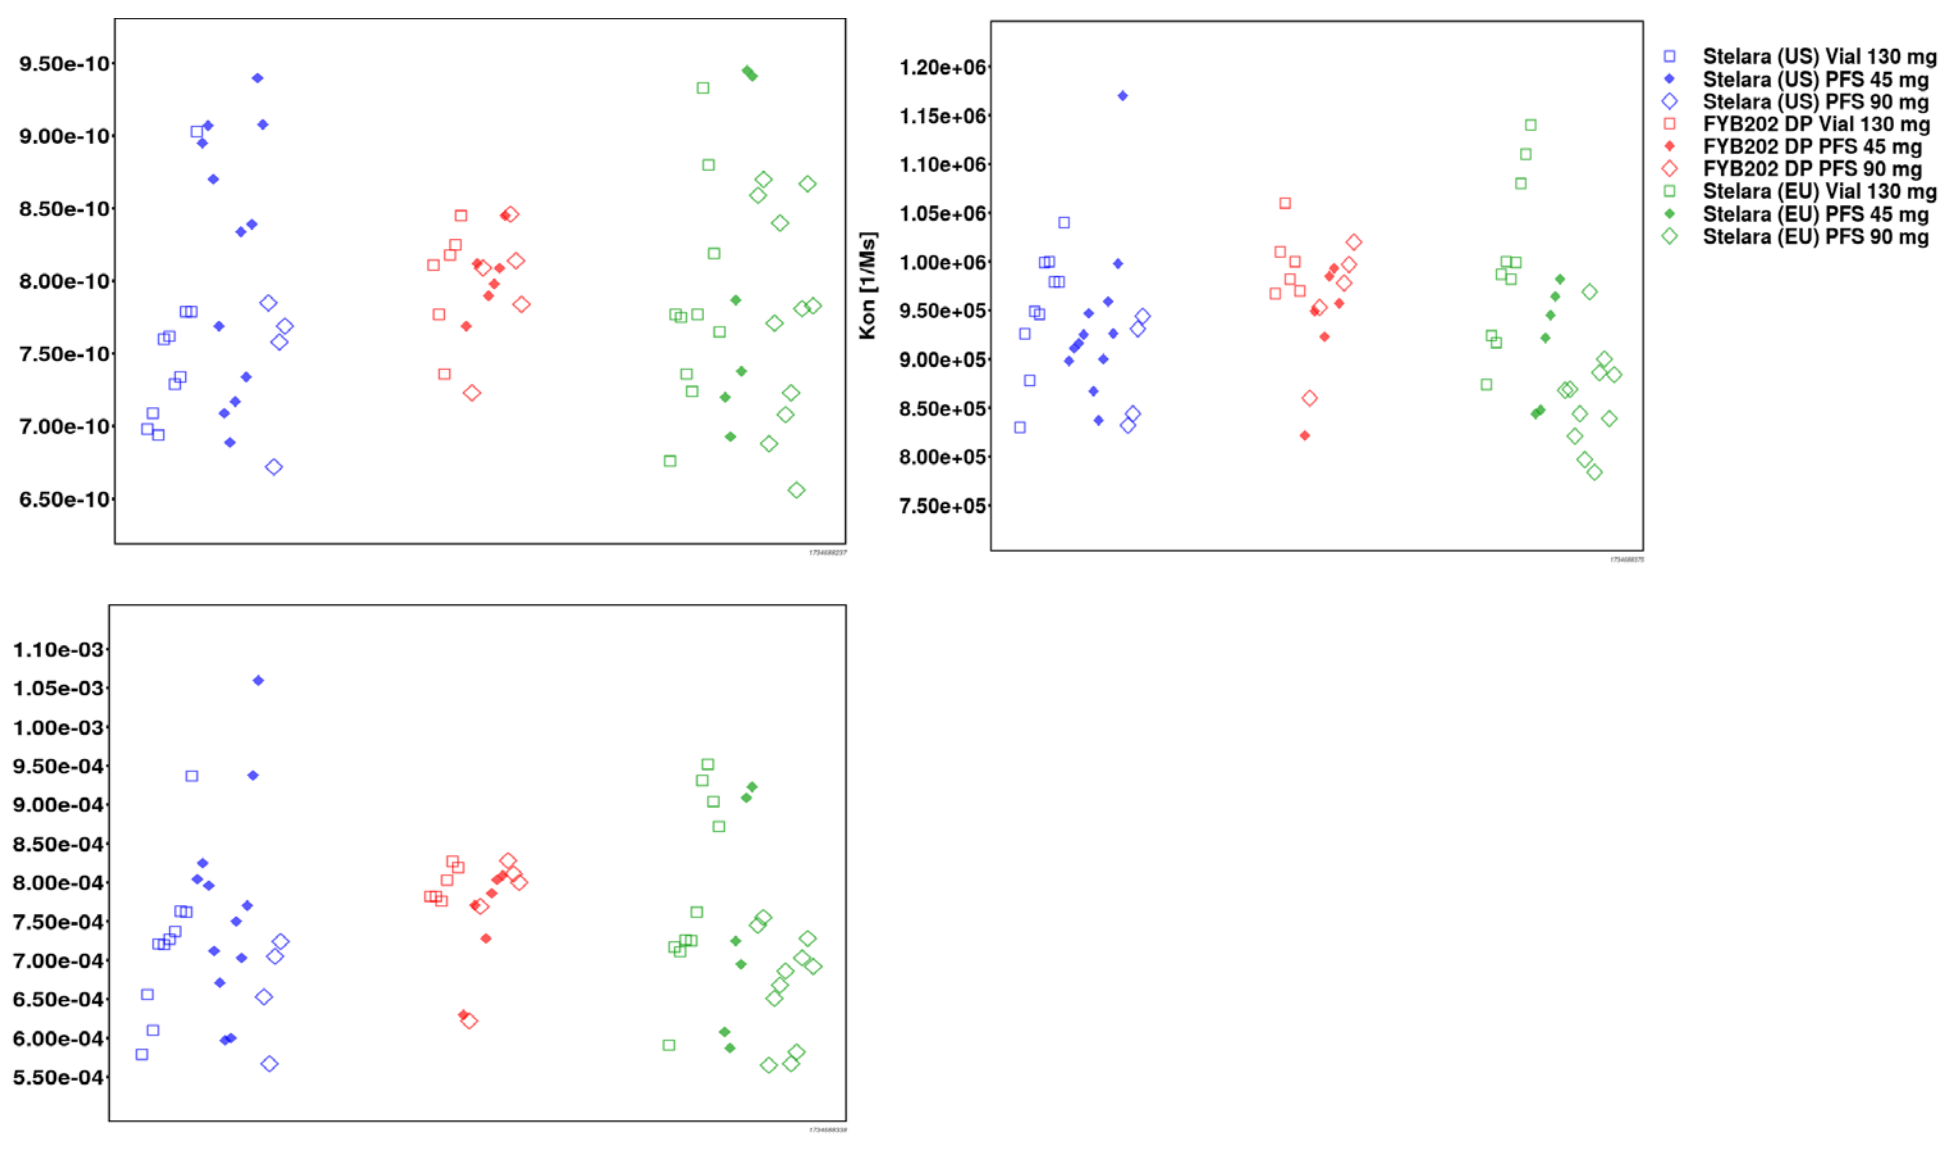
KD [M]
Kon [1/Ms]
Koff [1/s]
Stelara (US) Vial 130 mg
Stelara (US) PFS 45 mg
Stelara (US) PFS 90 mg
FYB202 DP Vial 130 mg
FYB202 DP PFS 45 mg
FYB202 DP PFS 90 mg
Stelara (EU) Vial 130 mg
Stelara (EU) PFS 45 mg
Stelara (EU) PFS 90 mg

- The **3-arm PK study** in 491 healthy volunteers demonstrated PK equivalence between FYB202 and both EU-approved and US-licensed ustekinumab and between the two reference drugs.
- The mean serum concentration-time profiles after a single subcutaneous dose of 45 mg ustekinumab were superimposable for all three treatments (Fig. 3).
- All 90% CIs were within the predefined equivalence margin of 80 to 125% (Balser[3]) (Fig. 4).

Figure 3: Superimposable PK profiles in healthy volunteers

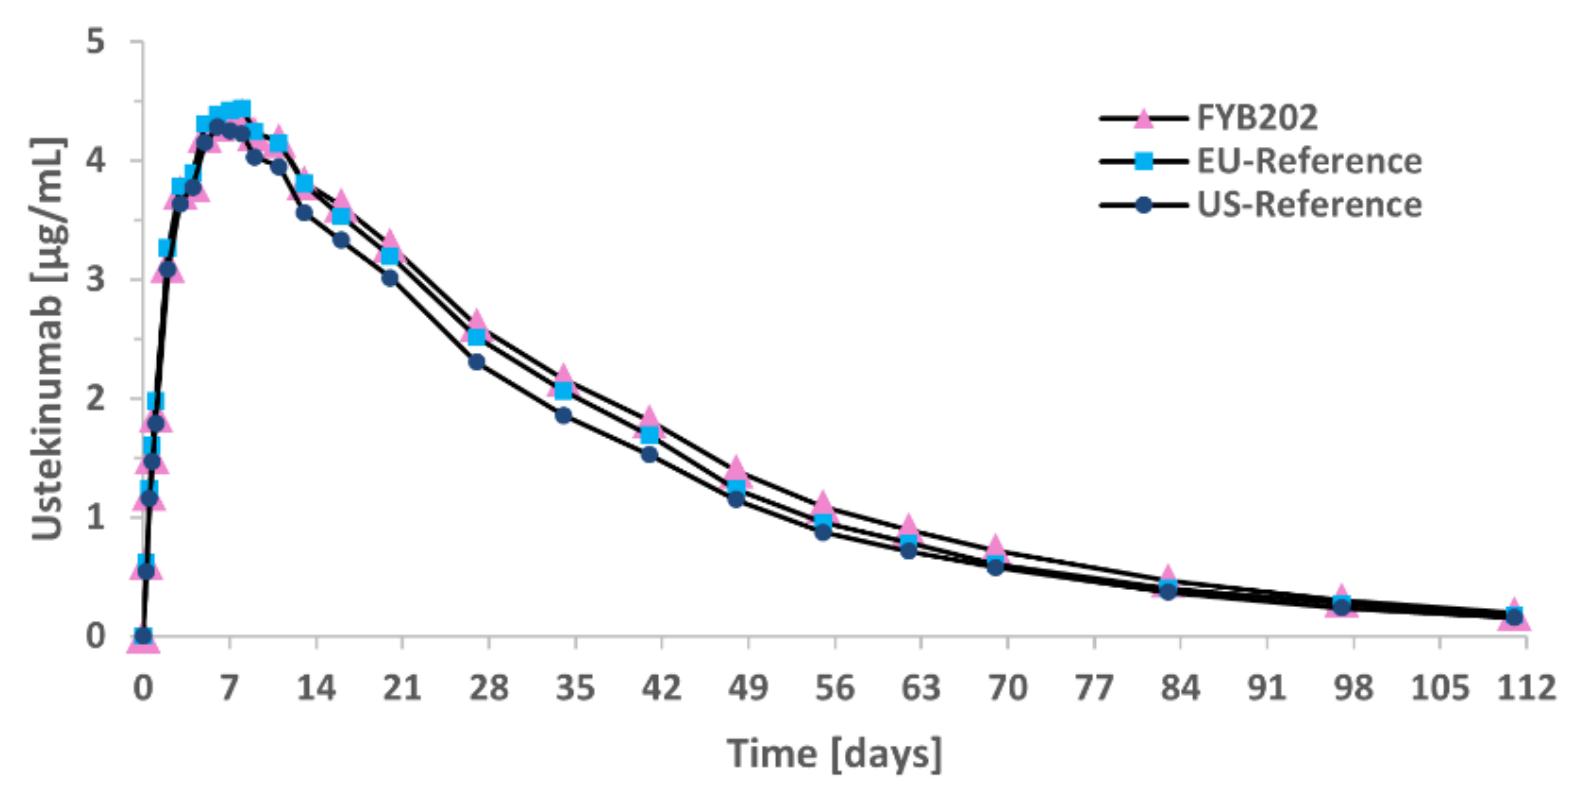
FYB202
EU-Reference
US-Reference
Ustekinumab [µg/mL]
Time [days]

Figure 4: Point estimates and 90% CIs for geom. mean ratios

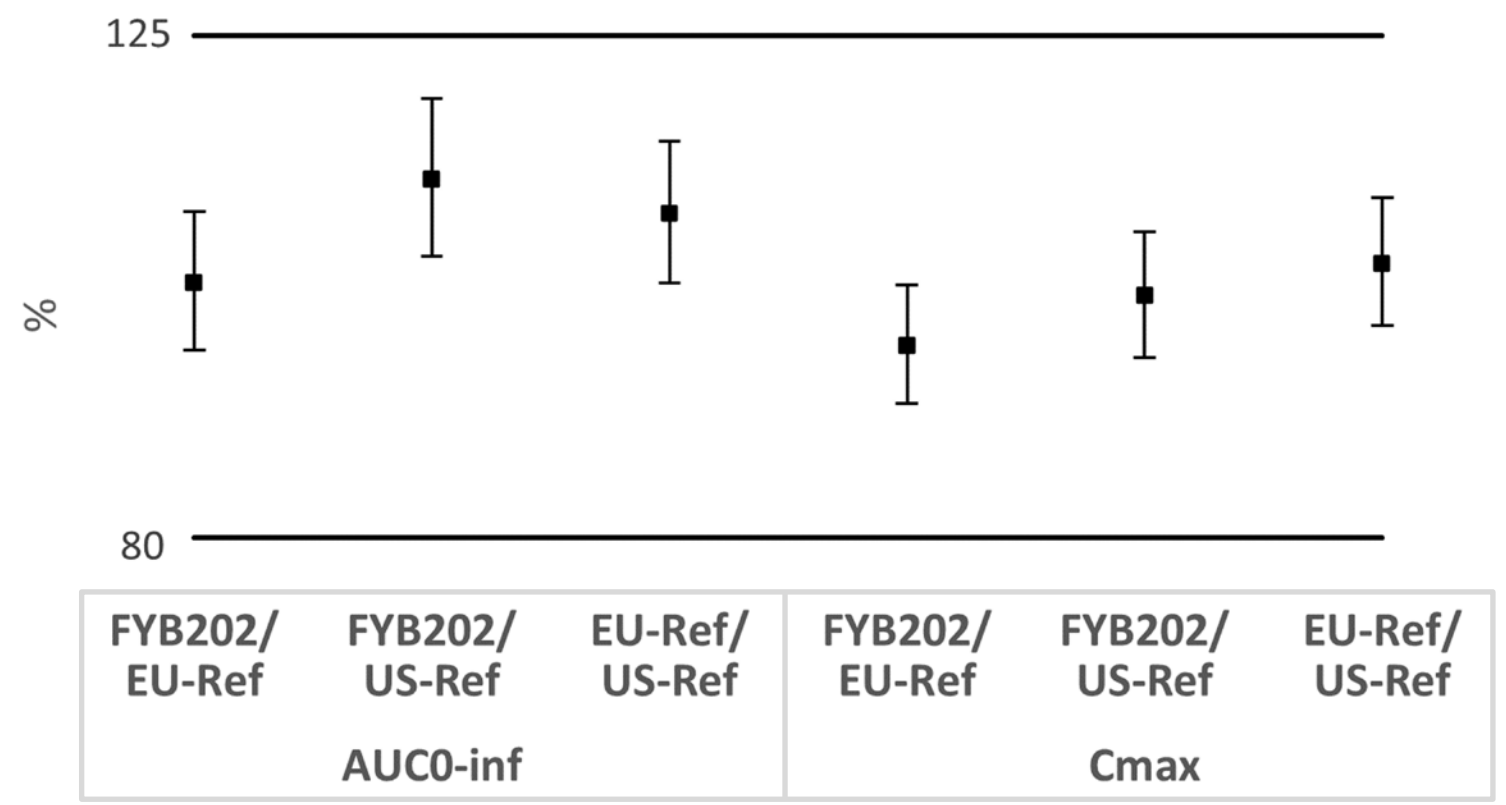
125
80
%
FYB202/ EU-Ref
FYB202/ US-Ref
EU-Ref/ US-Ref
AUC0-inf
Cmax

- After treatment, the percentage of subjects with at least one confirmed positive ADA and NAb result was lower in the FYB202 group compared to both reference groups (Fig. 5).

Figure 5: Percentage of healthy volunteers with at least one positive result

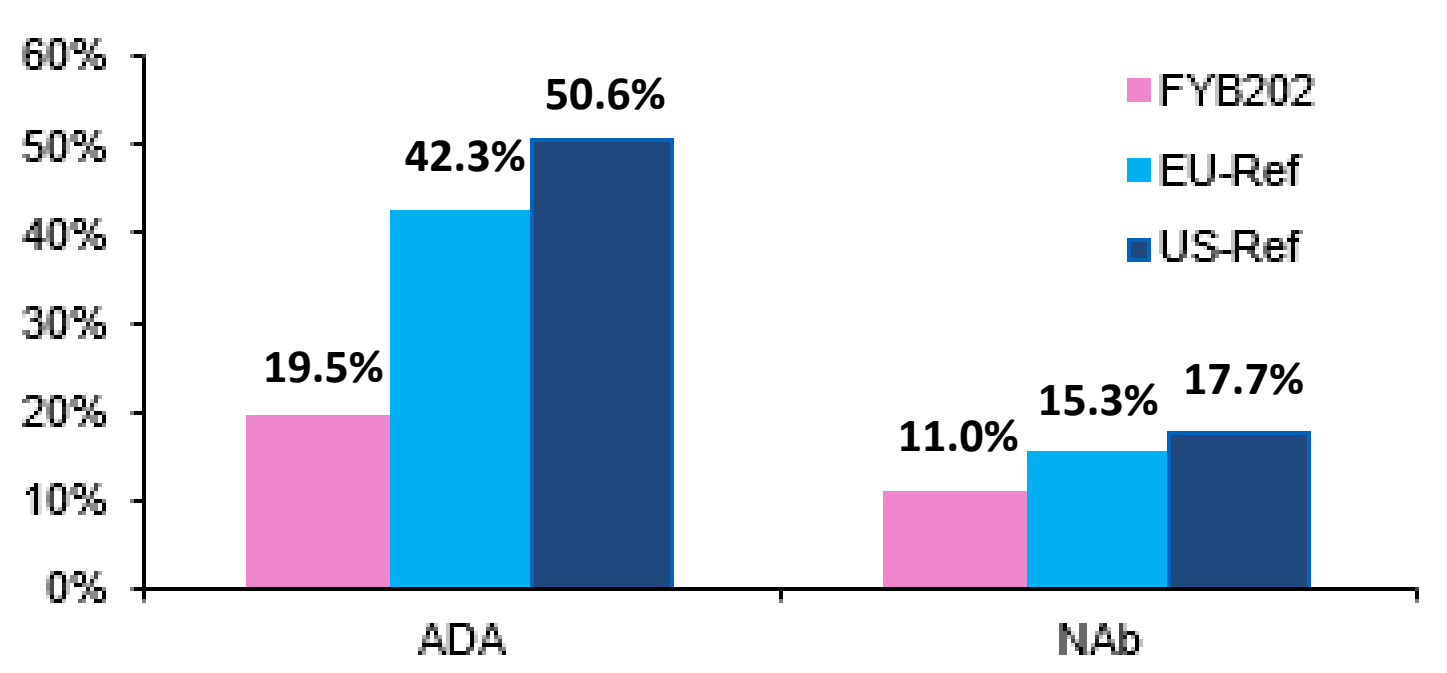
FYB202
EU-Ref
US-Ref
19.5%
42.3%
50.6%
11.0%
15.3%
17.7%
ADA
NAb

- For the primary endpoint in the **patient study**, estimated mean square differences together with the 90% and 95% CIs were fully contained within the respective equivalence margins (Papp[4]) (Fig. 6).

Figure 6. Equivalence testing results for % change from baseline to week 12 in PASI

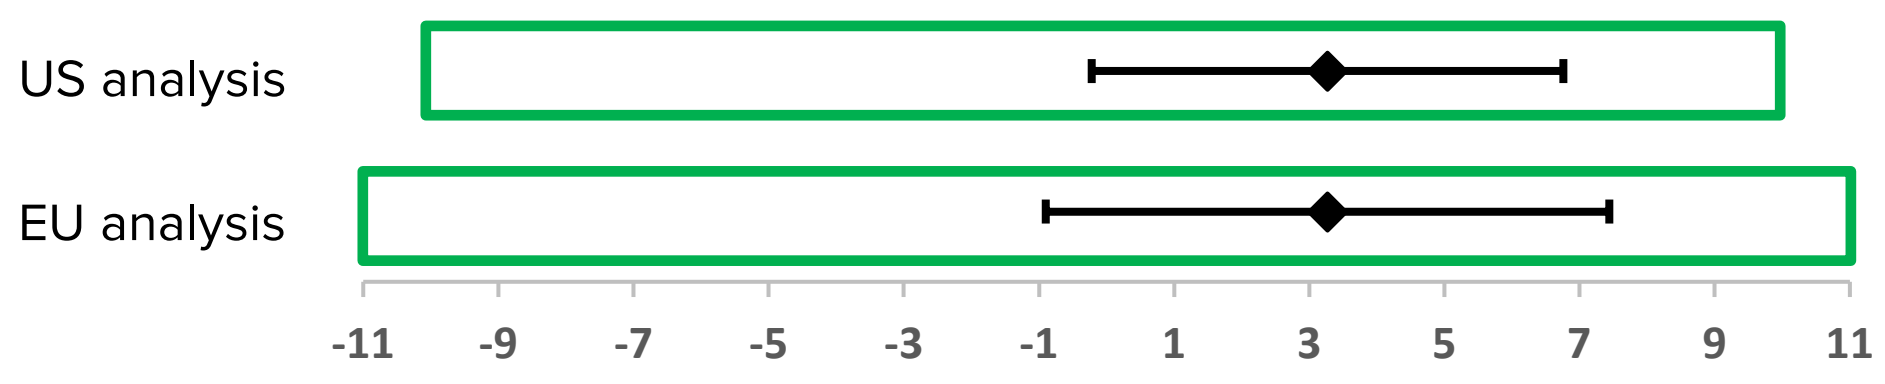
US analysis
EU analysis

- PASI75 responses over 52 weeks were highly similar in patients treated with FYB202 and reference ustekinumab and also stayed stable in patients switched from FYB202 to reference ustekinumab after 28 weeks (Fig. 7).

Figure 7. PASI75 response over 52 weeks

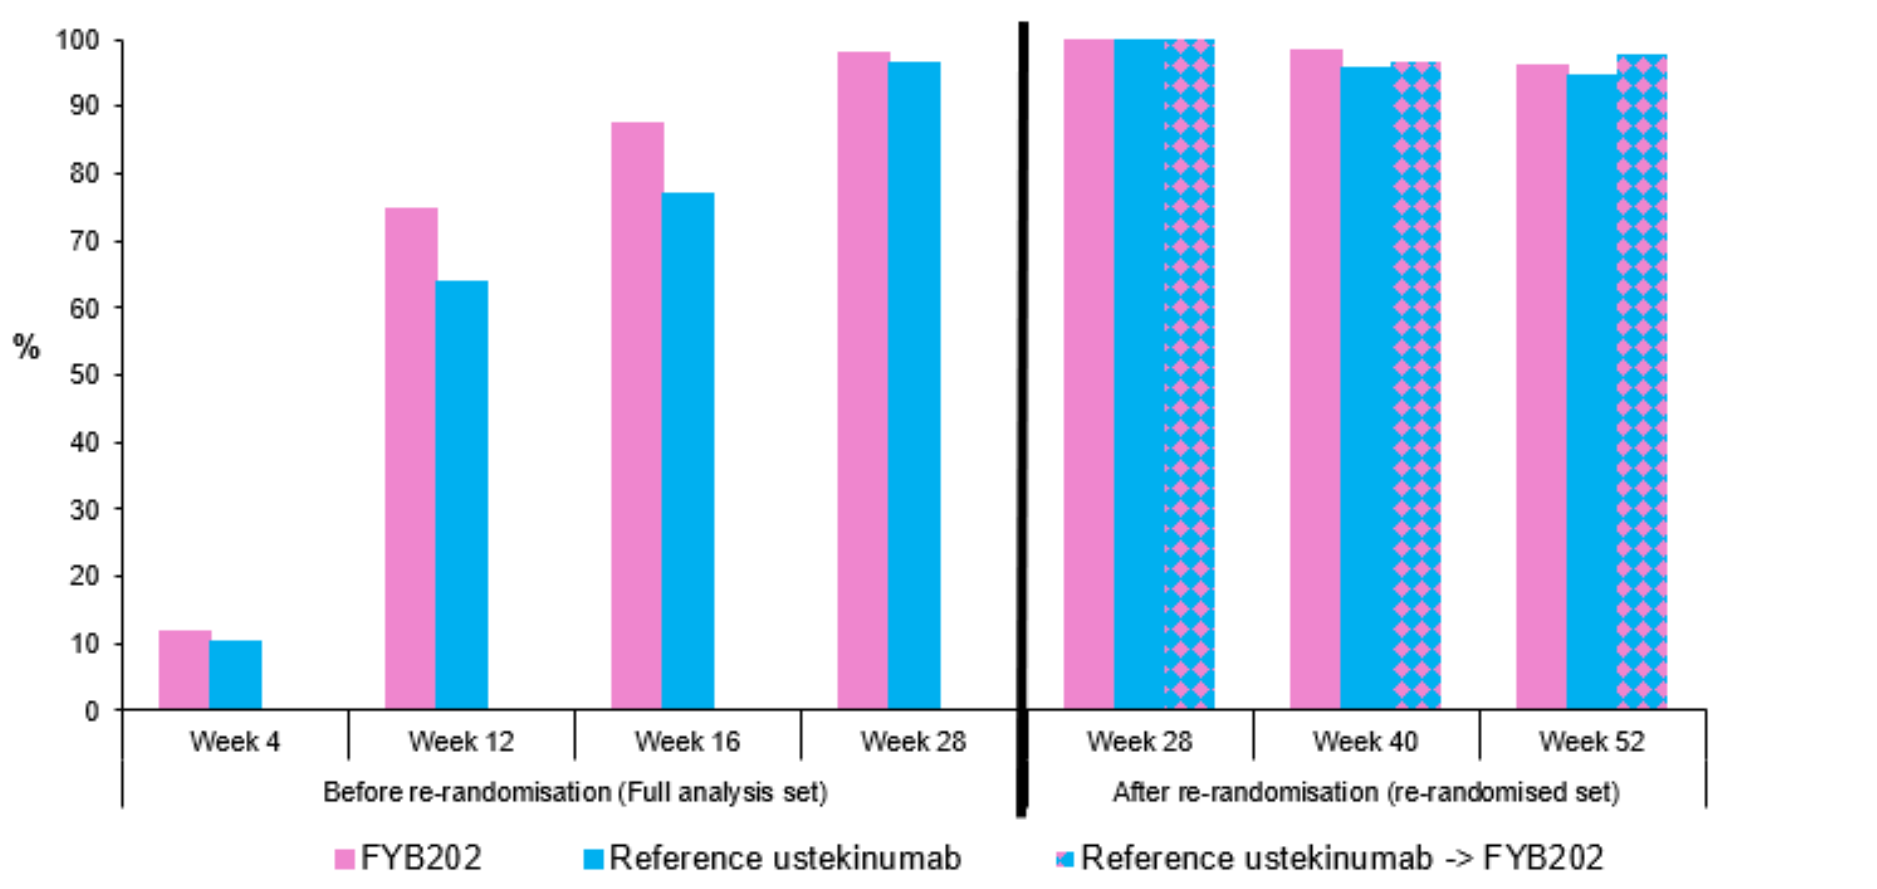
%
Week 4
Week 12
Week 16
Week 28
Week 28
Week 40
Week 52
Before re-randomisation (Full analysis set)
After re-randomisation (re-randomised set)
FYB202
Reference ustekinumab
Reference ustekinumab -> FYB202

- Incidences of ADAs and NAbs were lower in FYB202 treated patients (Fig. 8).
- No ADAs developed after transition from FYB202 to reference ustekinumab.

Figure 8: Percentage of patients with at least one positive result over 52 weeks

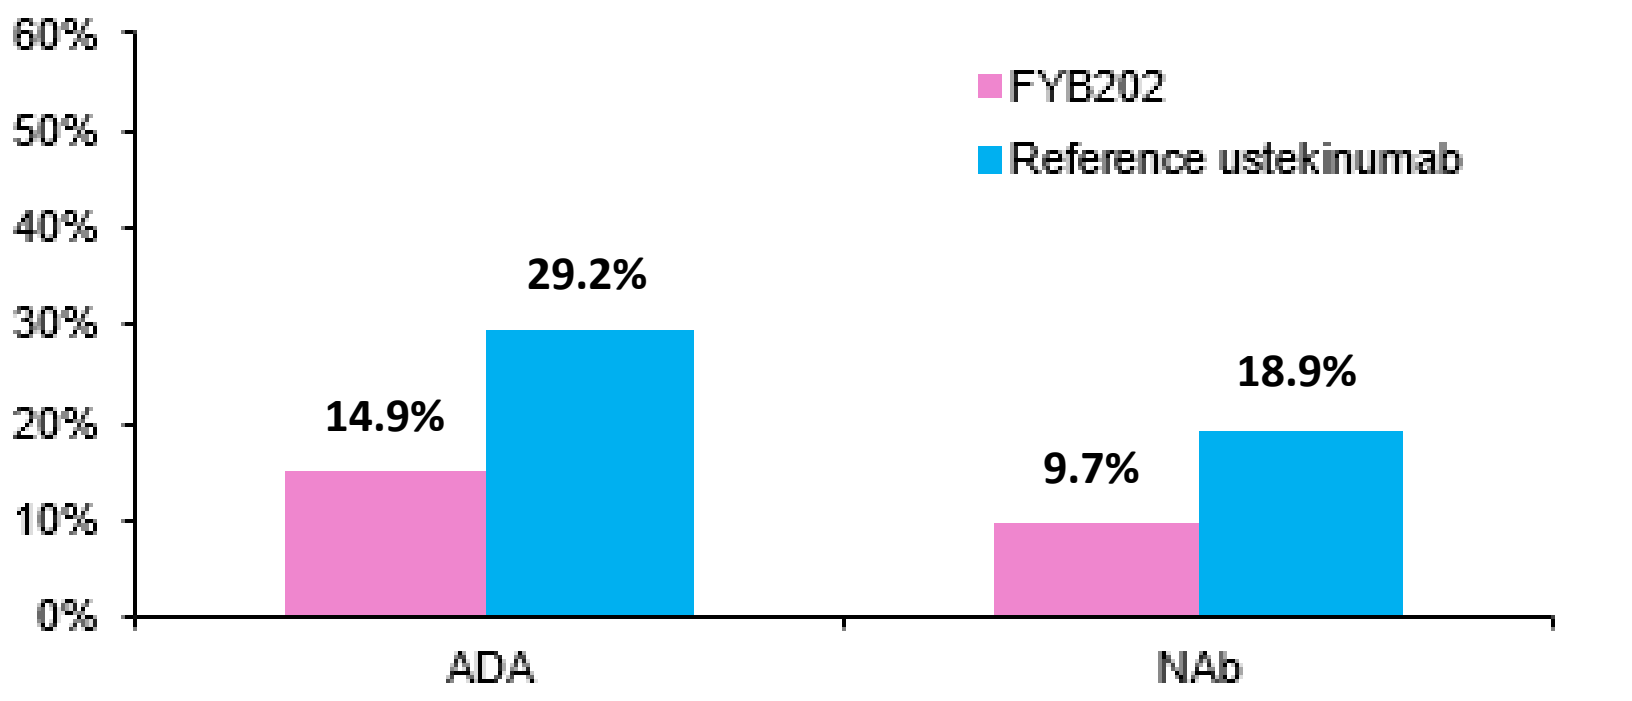
FYB202
Reference ustekinumab
14.9%
29.2%
9.7%
18.9%
ADA
NAb

References

1. Girolomoni G, *et al. Dermatol Ther (Heidelb*). 2023;13:2171–2185.
2. European Medicines Agency. Stelara: Product Information. https://www.ema.europa.eu/en/medicines/human/EPAR/stelara. Accessed August 2024
3. Balser, S., Nopora, K., Körner, J., Wedemeyer, R.-S., Anschütz, M. and Schug, B. (2024), New Ustekinumab Biosimilar Candidate FYB202: Pharmacokinetic Equivalence Demonstrated in a Randomized, Double-Blind, Parallel-Group, Single-Dose Trial in Healthy Subjects. Clin Pharmacol Drug Dev. https://doi.org/10.1002/cpdd.1473
4. Papp, K., Balser, S., Nopora, K., Freudensprung, B., Trieb, M. (2024), A randomised, double-blind trial to compare the efficacy, safety, and immunogenicity of the proposed biosimilar ustekinumab (FYB202) with reference ustekinumab in patients with moderate-to-severe plaque psoriasis. EADV 2024, Poster PO984

Conclusions

- FYB202 exhibits high analytical similarity to both EU- and US-reference products.
- The PK profiles of FYB202 and reference ustekinumab were superimposable and bioequivalence was established.
- FYB202 was shown to be equivalent to reference ustekinumab in patients with moderate-to-severe plaque psoriasis, with the mean percent improvement in PASI score from baseline to week 12 within the predefined equivalence intervals for both the EU- and US-specific analyses.
- After treatment, the incidences of confirmed positive ADA results were consistently lower in the FYB202 group compared to reference ustekinumab.
- Efficacy was maintained after switching from reference ustekinumab to FYB202 and switching had no clinically relevant effect on safety or immunogenicity.
- The totality of evidence, the consistent mode of action across the approved indications as well as the well-understood immunogenicity and safety profiles provide scientific justification for the safe and efficacious use of FYB202 in all approved clinical indications.

Funding and disclosures: The trials were originally funded by bioeq GmbH, which has since been acquired by Formycon AG.
FYB202 will be commercialized by Fresenius Kabi in partnership with Formycon AG. SB, BF, KH, IM, KN, SP, MS, MT, and FW are employees of Formycon AG.
Acknowledgements: The authors would like to thank the patients and study personnel involved in this study.